VIVIAD, A Phase 2b Study Investigating Varoglutamstat in Patients with MCI and Mild AD: Update on Dose Selection and Interim Blinded Safety Results

Frank Weber[1], Katharina Fuchs[1], Michael Schaeffer[1], Asger Reinstrup Bihlet[3], Peter Alexandersen[3], Tobias Melton Axelsen[5], Philip Scheltens[2], Everard Vijverberg[2]

[1] Vivoryon Therapeutics N.V., Halle and Munich, Germany; [2] Alzheimer Center Amsterdam, Vrije Universiteit Amsterdam, Amsterdam UMC, Amsterdam, The Netherlands; [3] NBCD A/S, Herlev, Denmark; [4] Sanos Clinic, Vejle, Denmark; [5] Sanos Clinic, Herlev, Denmark

Varoglutamstat Background

Varoglutamstat (PQ912), a small molecule glutaminyl cyclase (QPCT) inhibitor, reduces the brain levels of pyroglutamate-3-Abeta (N3pE-Abeta), a toxic Abeta variant shown to play a pivotal role in the development and progression of Alzheimer's disease (AD)[1]. Varoglutamstat is in clinical development as oral disease-modifying therapy.

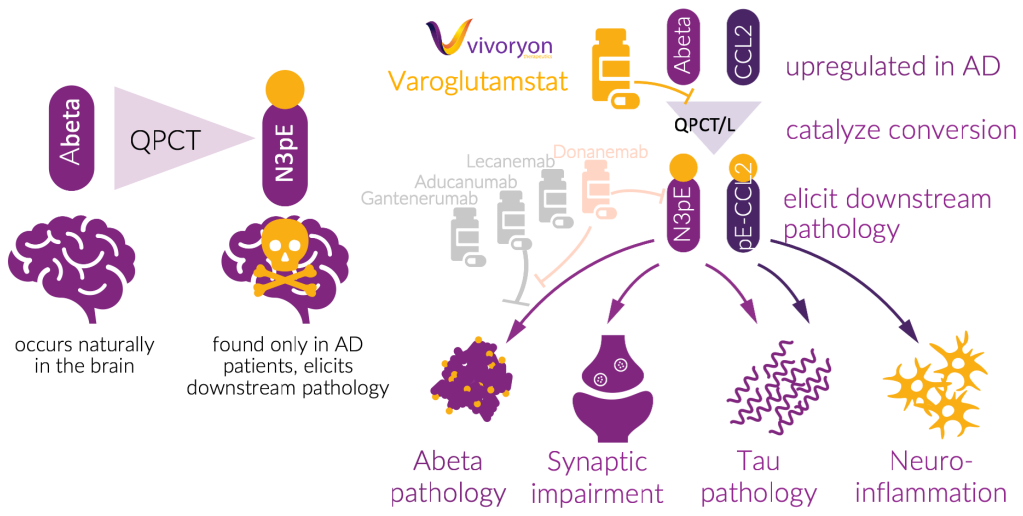
Abeta
QPCT
N3pE
occurs naturally in the brain
found only in AD patients, elicits downstream pathology
vivoryon
Varoglutamstat
Abeta
CCL2
QPCT/L
Lecanemab
Donanemab
Aducanumab
Gantenerumab
N3pE
pE-CCL2
upregulated in AD
catalyze conversion
elicit downstream pathology
Abeta pathology
Synaptic impairment
Tau pathology
Neuro-inflammation

- small molecule designed to target AD pathology upstream of Abeta-antibody approaches
- targets N3pE Abeta
- shown to impact synaptic impairment and tau pathology
- inhibits the full CCL2 maturation and modulates pro-inflammatory signaling

A prior Phase 2a study (NCT02389413) reported encouraging first evidence of the disease-modifying activity of varoglutamstat, most importantly with statistically significant changes from baseline in working memory as an important cognitive ability after only 12 weeks of treatment. While varoglutamstat was generally well tolerated, an MTD was reached at 800 mg twice daily (BID)[2].

These results led to the initiation of a state-of-the-art Phase 2b trial investigating multiple cognitive, safety and biomarker endpoints. Safety data reported to date show no on-target toxicity and no clinical signs of amyloid-related imaging abnormalities (ARIA), a severe side effect reported for antibody-based AD therapies.

Methods

VIVIAD (NCT04498650) is a multicenter randomized, placebo-controlled, double-blind, parallel group dose finding Phase 2b study in patients with early Alzheimer's disease (MCI due to AD and mild AD)[3].

The treatment duration varies between 48 and 96 weeks dependent on the time of inclusion. Efficacy is assessed by the Cogstate NTB and the Amsterdam Quality of Life Questionnaire (A-IADL-Q). Secondary endpoints include functional read-outs by EEG and assessment of functional and inflammatory biomarkers. In June 2022, an independent DSMB decided that the highest dose tested, 600 mg BID, a dose known to result in a target occupancy of close to 90%, was well tolerated and safe to be carried forward for the second part of the study. Patients previously randomized to 300 mg BID have been blindly up-titrated to 600 mg BID. All data remain blinded outside the DSMB.

(1) Schilling et al., Nat Med. 14, 1106 (2008). https://doi.org/doi: 10.1038/nm.1872
(2) Scheltens et al., Alz Res Therapy 10, 107 (2018). https://doi.org/10.1186/s13195-018-0431-6
(3) Vijverberg et al., Alz Res Therapy 13, 142 (2021). https://doi.org/10.1186/s13195-021-00882-9

VIVIAD Objectives and Overview

Evaluate the safety and efficacy of varoglutamstat in patients with early AD and mild cognitive impairment (MCI).

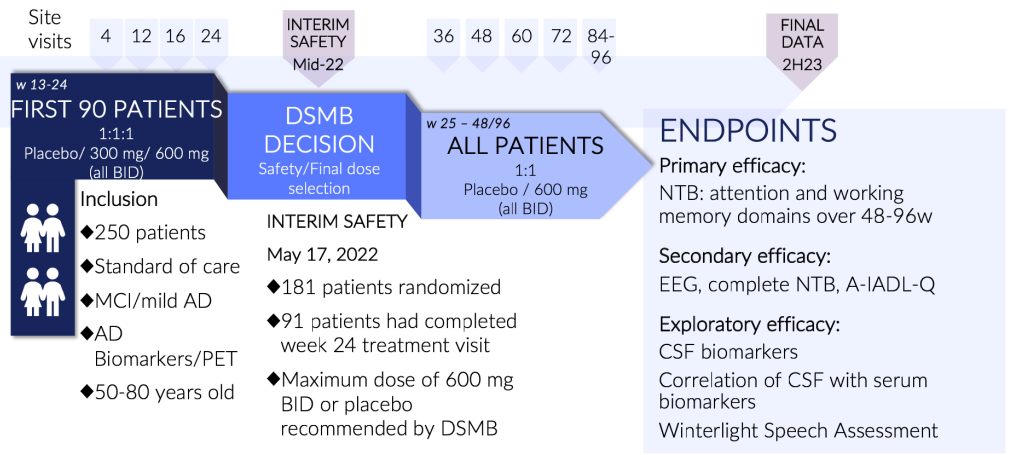
Site visits
4 12 16 24
INTERIM SAFETY Mid-22
36 48 60 72 84-96
FINAL DATA 2H23
w 13-24
FIRST 90 PATIENTS
1:1:1
Placebo/ 300 mg/ 600 mg (all BID)
DSMB DECISION
Safety/Final dose selection
w 25 – 48/96
ALL PATIENTS
1:1
Placebo / 600 mg (all BID)
Inclusion
◆250 patients
◆Standard of care
◆MCI/mild AD
◆AD Biomarkers/PET
◆50-80 years old
INTERIM SAFETY
May 17, 2022
◆181 patients randomized
◆91 patients had completed week 24 treatment visit
◆Maximum dose of 600 mg BID or placebo recommended by DSMB
ENDPOINTS
Primary efficacy:
NTB: attention and working memory domains over 48-96w
Secondary efficacy:
EEG, complete NTB, A-IADL-Q
Exploratory efficacy:
CSF biomarkers
Correlation of CSF with serum biomarkers
Winterlight Speech Assessment

The study is enrolling patients at 22 AD study centers in five European countries, DK, NL, DE, ES, PL, and will randomize 250 patients.

Baseline Data (Blinded Data)

Data from 224 (CSF: N=220) randomized patients	Min	Max	Mean	Median	SD
Age (years)*	50	80	68.3	70.0	7.4
DSST^	0	72	31.3	31.0	15.4
MMSE*	20	30	24.4	24.5	2.7
A-IADL-Q*	25.5	73.6	51.0	50.5	8.6
Amyloid Beta 1-42 (pg/mL)*	237	1377	659.8	639.0	221.9
Phosphorylated Tau Protein (pg/mL)*	9	108.7	35.2	32.8	15,3
P-TAU/AMYLB42 (ratio)	0.007	0.218	0.057	0.052	0.029

Genders are close to equally distributed with 51 % female and 49 % male patients with an average age of 68 years.

MMSE: Mini-Mental State Examination DSST: Digital Symbol Substitution Test
A-IADL-Q: Amsterdam Instrumental Activities of Daily Living Questionnaire
* collected at screening ^collected at baseline, N=221 SD = Standard Deviation

Duration of patients in study (Oct 7, 2022)

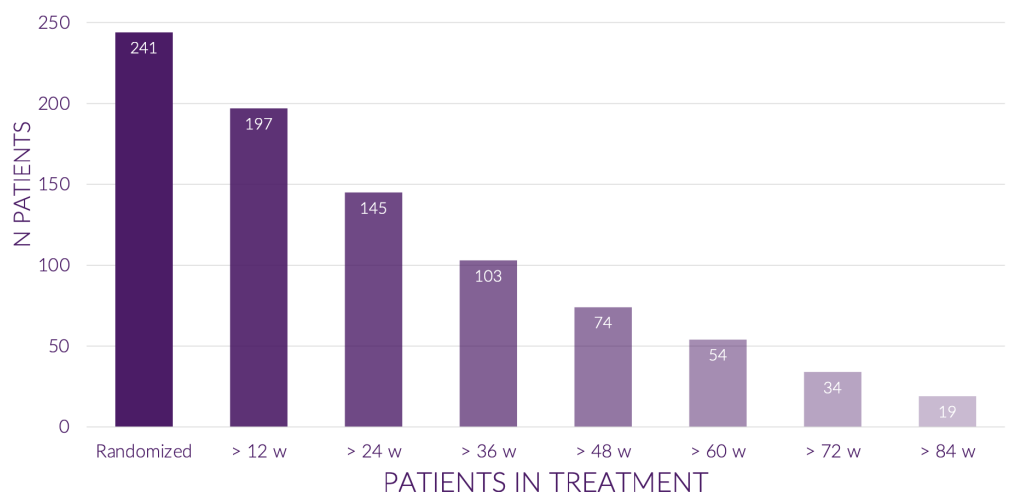
N PATIENTS
250
200
150
100
50
0
241
197
145
103
74
54
34
19
Randomized
> 12 w
> 24 w
> 36 w
> 48 w
> 60 w
> 72 w
> 84 w
PATIENTS IN TREATMENT

Interim Safety Results (Blinded Data)

As of October 7, 2022, the study has enrolled 241 patients, the total number planned is 250. At this point in time 400 patients were screen failures which corresponds to 62 %.

As recommended by the DSMB, all patients are dosed with either 600 mg BID varoglutamstat or placebo. The occurrence of adverse events normalized per 100 visits continues to be stable around 31, indicating that up-titration from 300 to 600 mg twice daily did not result in an increased frequency of adverse events.

- only 17% of AEs were assessed as potentially related to the study treatment, only one of these was severe
- the majority of the related AEs belong to gastrointestinal disorders.
- there were no on-study deaths.
- most TEAEs were mild (70%) or moderate (28%).
- no clinical signs of ARIAs were observed.

Overall Summary of Adverse Events (AE) Oct 7, 2022	Total (N=241) n (%) E
Any Adverse Events (AEs)	173 (71.7) 480
Treatment Emergent Adverse Events (TEAEs)	142 (58.9) 401
Deaths	0 (0.0) 0
Serious Adverse Events (SAEs)	9 (3.7) 12
AEs leading to discontinuation from the study	4 (1.7) 4
Amyloid-related imaging abnormalities (ARIAs)	0 (0.0) 0

Treatment Emergent AEs

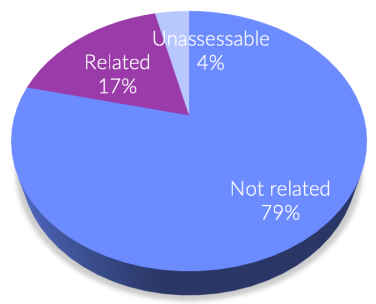
Unassessable 4%
Related 17%
Not related 79%

Severity of TEAEs

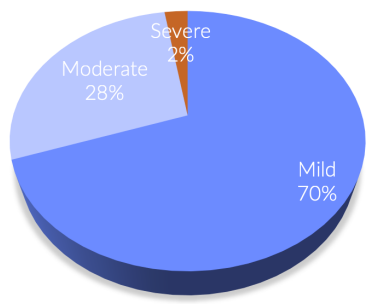
Severe 2%
Moderate 28%
Mild 70%

TEAEs by SOC

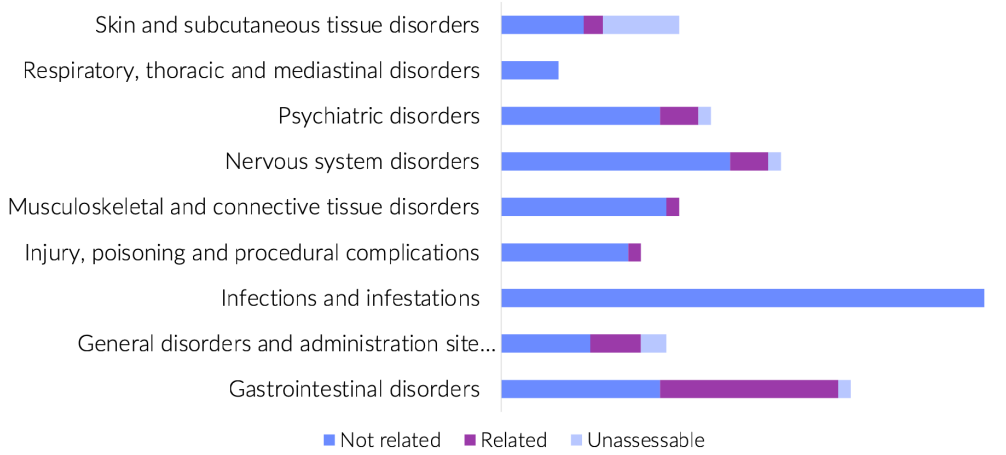
Skin and subcutaneous tissue disorders
Respiratory, thoracic and mediastinal disorders
Psychiatric disorders
Nervous system disorders
Musculoskeletal and connective tissue disorders
Injury, poisoning and procedural complications
Infections and infestations
General disorders and administration site…
Gastrointestinal disorders
Not related
Related
Unassessable

TEAE: Treatment emergent adverse event SOC: system organ class

Conclusions

The state-of-the art Phase 2b study VIVIAD aims to yield important results in early AD for varoglutamstat, the first small molecule and only project in clinical development selectively targeting the *de novo* production of neurotoxic N3pE-Abeta.

Through a carefully crafted study design, the study was able to achieve improved tolerability for varoglutamstat compared to a prior Phase 2a study (NCT02389413), without significantly sacrificing target engagement. After the DSMB decision, the study continues as planned and patients will receive 600 mg BID active or placebo for at least 48 weeks.